ANTI-ABETA LIPOSOMAL VACCINE, ACI-24.060, RETAINS MEMORY IN AN AGGRESSIVE MOUSE MODEL OF ALZHEIMER'S DISEASE

Poster P0215 / #1422

Emma Fiorini, Chiara Babolin, Catherine Morici, Aurelie Guillaume Gentil, Fabiana Tione, Harishini Balachandiran, Maxime Ayer, Marie-Gabrielle Beuzelin, Rakel Carpintero, Stefania Rigotti, Andrea Pfeifer, Marie Kosco-Vilbois and Marija Vukicevic - AC Immune SA - Lausanne (Switzerland)

AC Immune

Introduction

ACI-24.060 is a Phase 2 clinical stage anti-Abeta active immunotherapy that, in preclinical studies, induces sustained antibody responses targeting pathological Abeta species (Vukicevic, 2022). Here, we report the ability of ACI-24.060 to maintain neuronal health using a cognitive test for learning and memory in the 5xFAD model of Alzheimer's disease.

SupraAntigen® platform

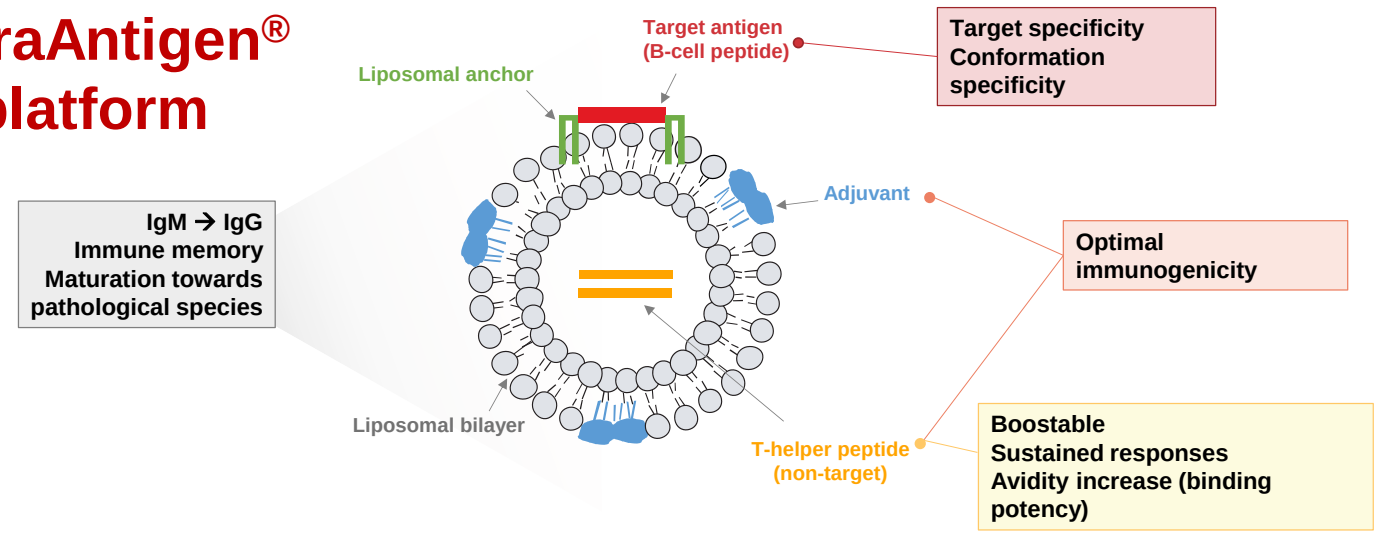
Target antigen (B-cell peptide)
Target specificity
Conformation specificity
Liposomal anchor
Adjuvant
Optimal immunogenicity
IgM → IgG
Immune memory
Maturation towards pathological species
Liposomal bilayer
T-helper peptide (non-target)
Boostable
Sustained responses
Avidity increase (binding potency)

Figure 1: *ACI-24.060 is an optimized liposomal SupraAntigen® vaccine formulation*

ACI-24.060 induced a strong and sustained polyclonal response in 5xFAD mice

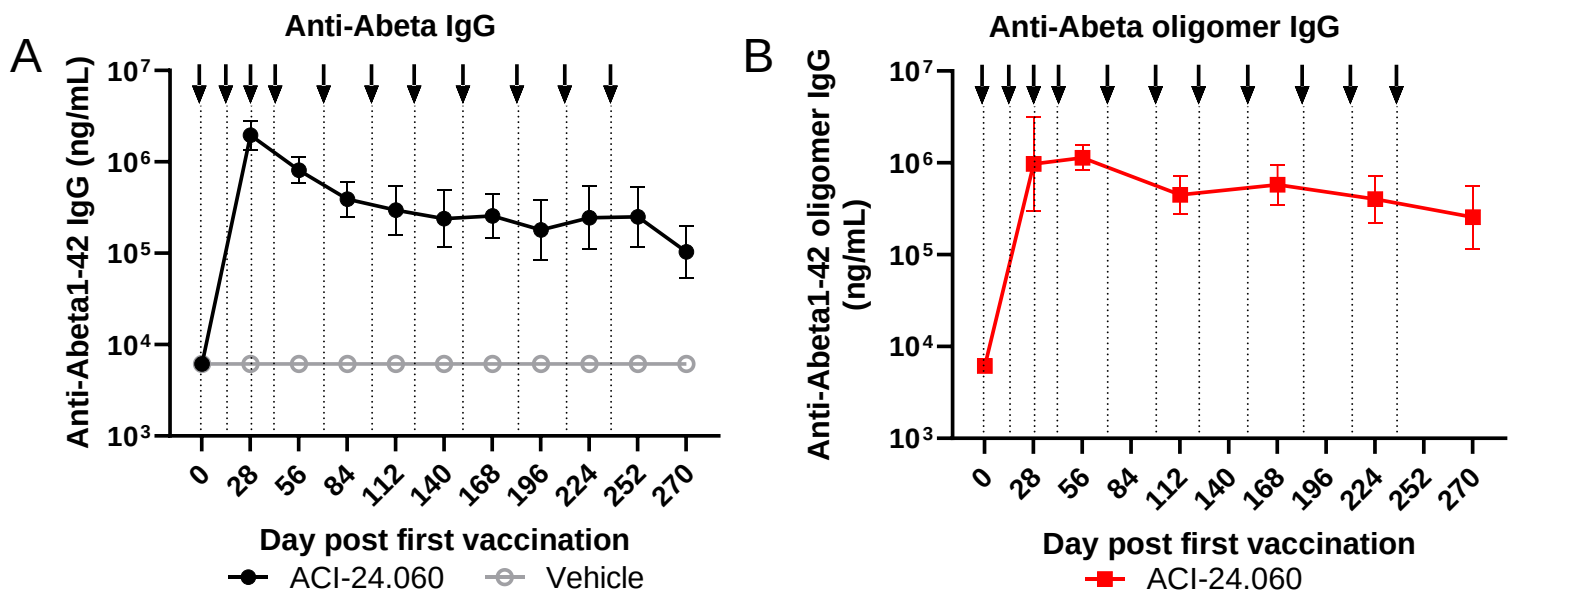
A
Anti-Abeta IgG
Anti-Abeta1-42 IgG (ng/mL)
10^7
10^6
10^5
10^4
10^3
0 28 56 84 112 140 168 196 224 252 270
Day post first vaccination
ACI-24.060
Vehicle
B
Anti-Abeta oligomer IgG
Anti-Abeta1-42 oligomer IgG (ng/mL)
Day post first vaccination
ACI-24.060

Figure 2: *Mice received eleven injections with ACI-24.060 (8µg/dose) or vehicle over a period of 9 months (arrows indicate injections), starting at 1.5 month of age (n=15/group). A) Anti-Abeta and B) anti-Abeta oligomer IgG in mouse plasma (n=15/group) at predose and postdose. Data are shown as geometric mean with 95% confidence interval (CI).*

ACI-24.060 induces antibodies with a 3-log preferential recognition of Abeta oligomers vs monomers

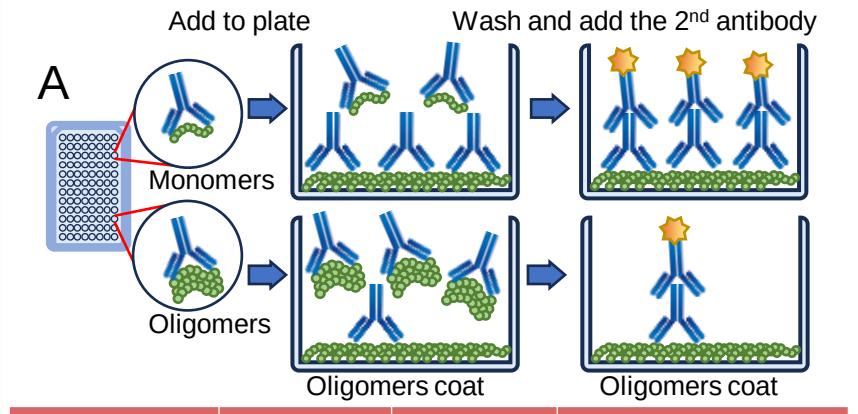
A
Add to plate
Wash and add the 2nd antibody
Monomers
Oligomers
Oligomers coat
Oligomers coat

Mouse plasma Day 252 (n=12)	Abeta monomers	Abeta oligomers	Ratio IC50 Abeta monomers/ Abeta oligomers
IC50 nM (Average)	463	0.14	3307

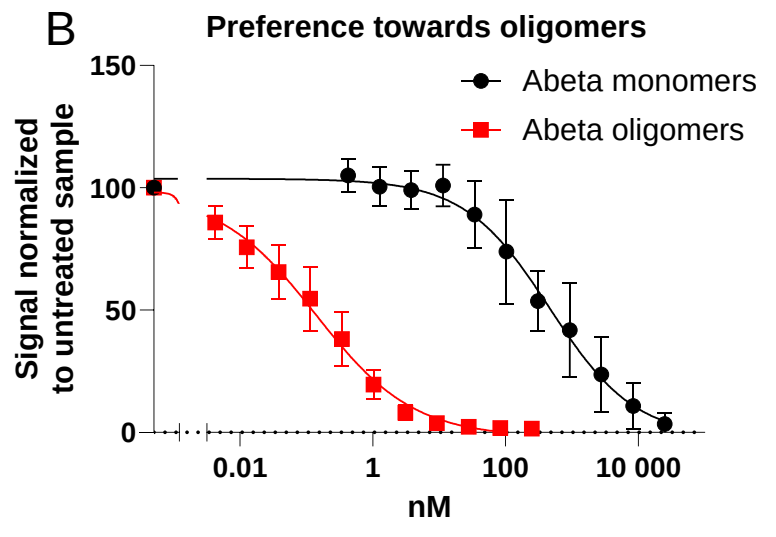
B
Preference towards oligomers
Abeta monomers
Abeta oligomers
Signal normalized to untreated sample
150
100
50
0
0.01
1
100
10 000
nM

Figure 3: *A) Scheme of the assay set-up (Soderberg, 2022). B) Mouse plasma (n= 12) (collected after 11 injections with ACI-24.060) at 1/200 were incubated with increasing concentration of Abeta oligomers (squares) or monomers (circles). The signal in the presence of Abeta species is normalized to the untreated sample. IC50 was calculated and used to obtain the monomer vs oligomer ratio. Graph shows the mean inhibition of individual mice ±SD.*

ACI-24.060 maintained both learning and memory capacities comparable to animals without disease

DAY	LEARNING TRIAL				PROBE TRIAL	Platform
	Trial 1	Trial 2	Trial 3	Trial 4		
1	SE	SW	NW	SW	-	NE
2	NW	SE	SW	NW	-	NE
3	SW	NW	SE	NW	-	NE
4	SE	SW	NW	SE	-	NE
5	-	-	-	-	SW	Removed

Quadrant:
SW: South-West
SE: South-East
NW: North-West
NE: North-East

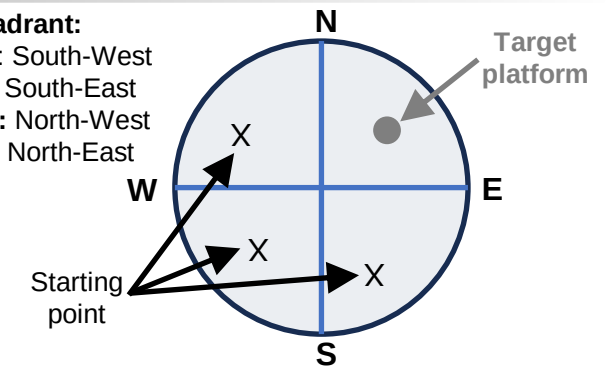
N
S
W
E
Target platform
Starting point
X
X
X

Figure 4*: Set up of the Morris-Water Maze Test: Mice were trained for four days to find the hidden platform (learning trial, Day 1-4) , and then on the fifth day the platform was removed, and the memory capacity was assessed (probe trial, Day 5).*

Learning capacity during training (learning trial)

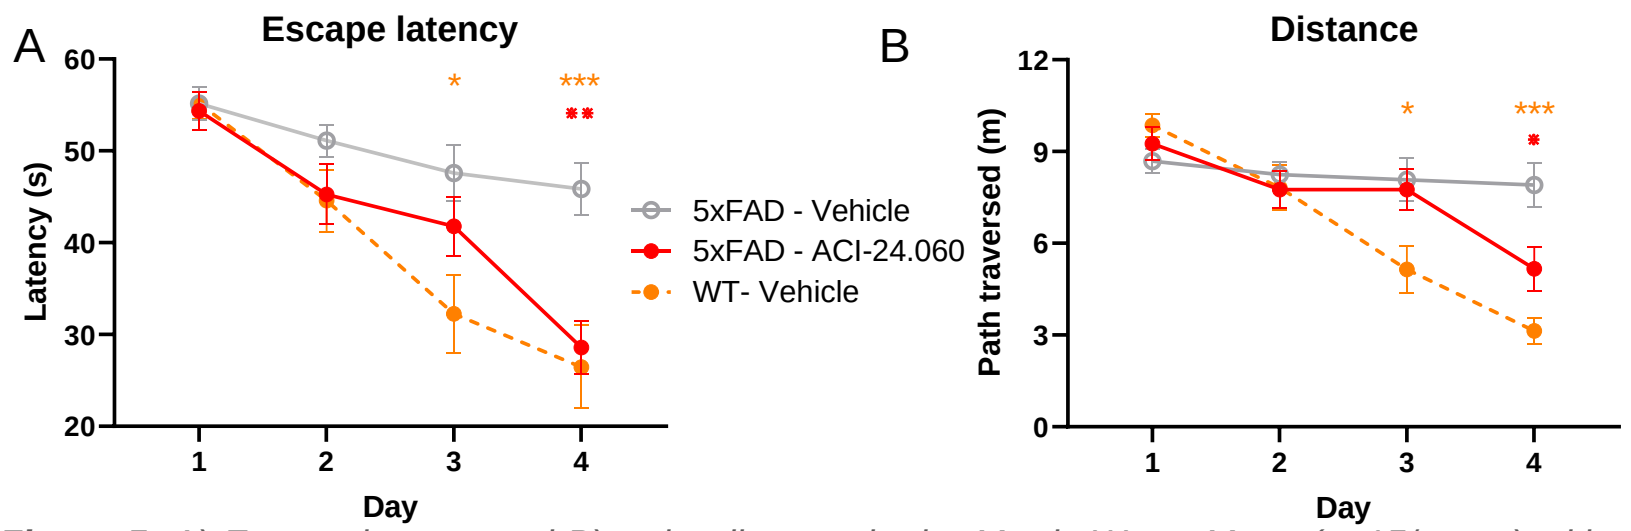
A
Escape latency
Latency (s)
60
50
40
30
20
1 2 3 4
Day
5xFAD - Vehicle
5xFAD - ACI-24.060
WT- Vehicle
B
Distance
Path traversed (m)
12
9
6
3
0
1 2 3 4
Day

Figure 5: *A) Escape latency and B) swim distance in the Morris Water Maze (n=15/group) with wild-type (WT) or 5xFAD mice at the age of ~11 months. Results are shown as group means ± SEM. Statistics: Repeated measures two-way ANOVA followed by Bonferroni's multiple comparisons test, using vehicle treated 5xFAD mice as a reference group. *(5xFAD - Vehicle vs. WT- Vehicle) or *(5xFAD - Vehicle vs. 5xFAD- ACI-24.060). */* p<0.05, **p<0.05, ***/*** p<0.001*

Post-training memory evaluation (probe trial)

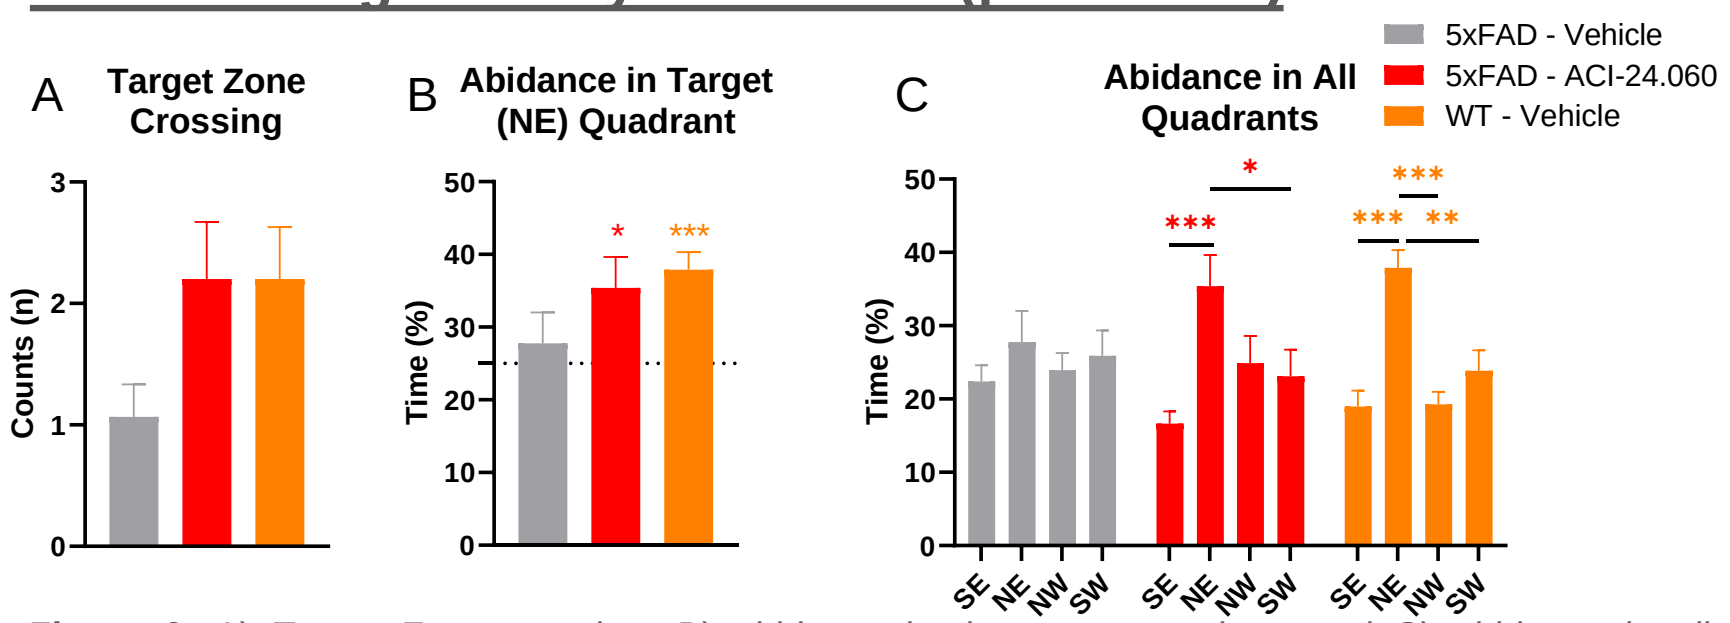
A
Target Zone Crossing
Counts (n)
B
Abidance in Target (NE) Quadrant
Time (%)
C
Abidance in All Quadrants
Time (%)
SE NE NW SW
5xFAD - Vehicle
5xFAD - ACI-24.060
WT - Vehicle

Figure 6*: A) Target Zone crossing, B) abidance in the target quadrant and C) abidance in all quadrants. Results are shown as group means ± SEM. Statistics: (A) Mann-Whitney tests; (B)One sample t-test against a random abidance in the target quadrant of 25%; (C) Two-way ANOVA followed by Bonferroni's multiple comparisons test (within each group). *p<0.05,**p<0.05, *** p<0.001*

Cognitive score of training and probe trial

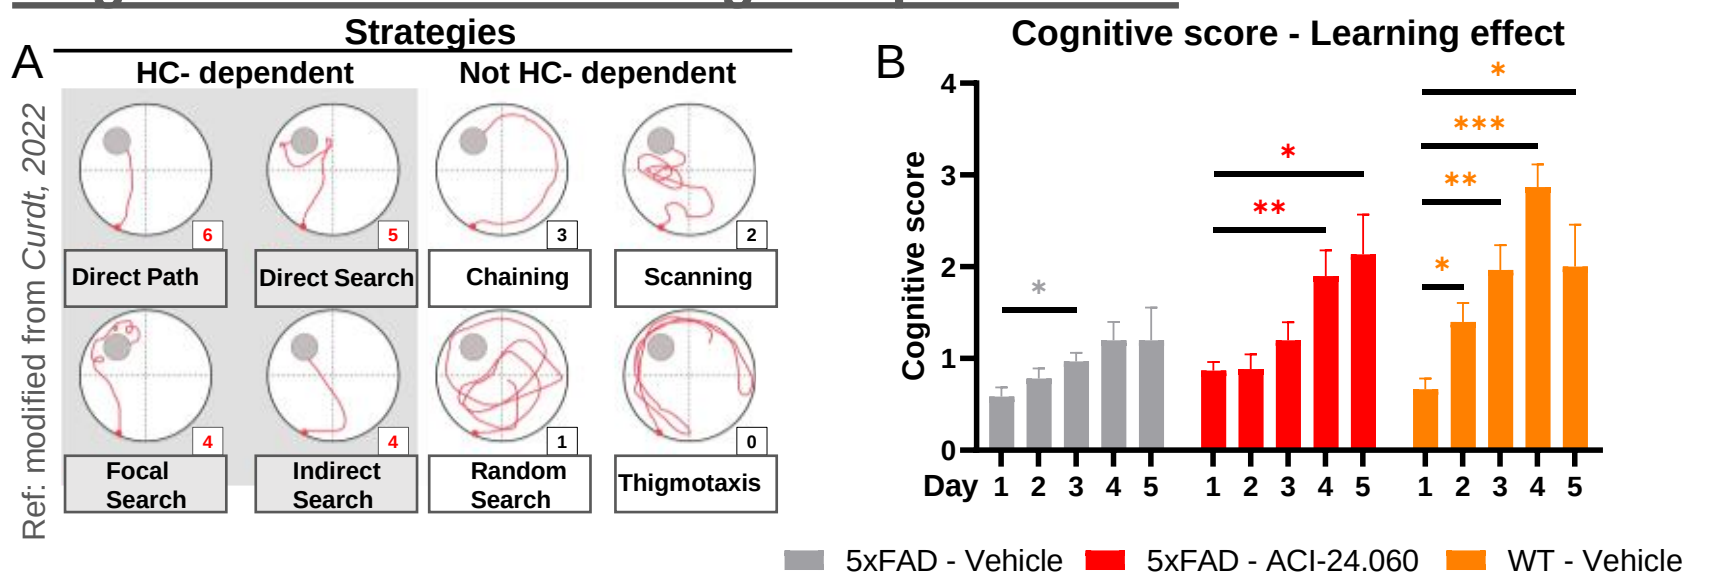
A
Strategies
HC- dependent
Not HC- dependent
Direct Path
Direct Search
Chaining
Scanning
Focal Search
Indirect Search
Random Search
Thigmotaxis
6
5
3
2
4
4
1
0
Ref: modified from Curdt, 2022
B
Cognitive score - Learning effect
Cognitive score
Day 1 2 3 4 5
5xFAD - Vehicle
5xFAD - ACI-24.060
WT - Vehicle

Figure 7*: A) Hippocampus (HC)-related cognitive scores in Water Maze test. B) Results are shown as group means ± SEM. Statistics: Repeated measures two-way ANOVA followed by Bonferroni's multiple comparisons test. * p<0.05, **p<0.05, *** p<0.001*

Conclusions

- ACI-24.060 induced antibody responses in an aggressive mouse model, 5xFAD, with robust and sustained titers against Abeta pathological oligomers
- ACI-24.060 immunization maintained:
 - ✓ Learning capacities at the levels of non-diseased animals
 - ✓ Memory capacities mirroring the performance of animals without disease
- The safety and efficacy of the ACI-24.060 vaccine is currently being evaluated in the clinical trial Abate (CT.gov # NCT05462106)

AC Immune, ADPD-2024, March 2024

© 2024 AC Immune. Not to be used or reproduced without permission