LP130

Anti-Abeta liposomal vaccine, ACI-24.060, induces anti-Abeta antibodies with binding profiles mirroring clinically validated monoclonal antibodies

Emma Fiorini, Chiara Babolin, Rakel Carpintero, Stefania Rigotti, Stefanie Siegert, Catherine Morici, Maëlle Verardo, Harishini Balachandiran, Jonathan Wagg, Piergiorgio Donati, Saskia Delpretti, Johannes Streffer, Andrea Pfeifer, Marie Kosco-Vilbois and Marija Vukicevic. AC Immune SA - Lausanne (Switzerland)

Background:

Active immunization is an attractive approach in development for the treatment and prevention of Alzheimer's disease (AD) and other neurodegenerative disorders. Its key advantage over other therapies is the induction of sustained antibody responses which can be easily boosted with occasional immunizations. The recent clinical evidence of targeting pathological species of amyloid with monoclonal antibodies (mAbs) such as lecanemab or donanemab, has validated the relevance of anti-Abeta immunotherapies. ACI-24.060 is a liposomal anti-Abeta vaccine, having Abeta 1-15 as immunogen. In this study in non-human primates (NHP), the antibodies produced post administration of ACI-24.060 were compared to the binding preferences towards different Abeta species of lecanemab and donanemab.

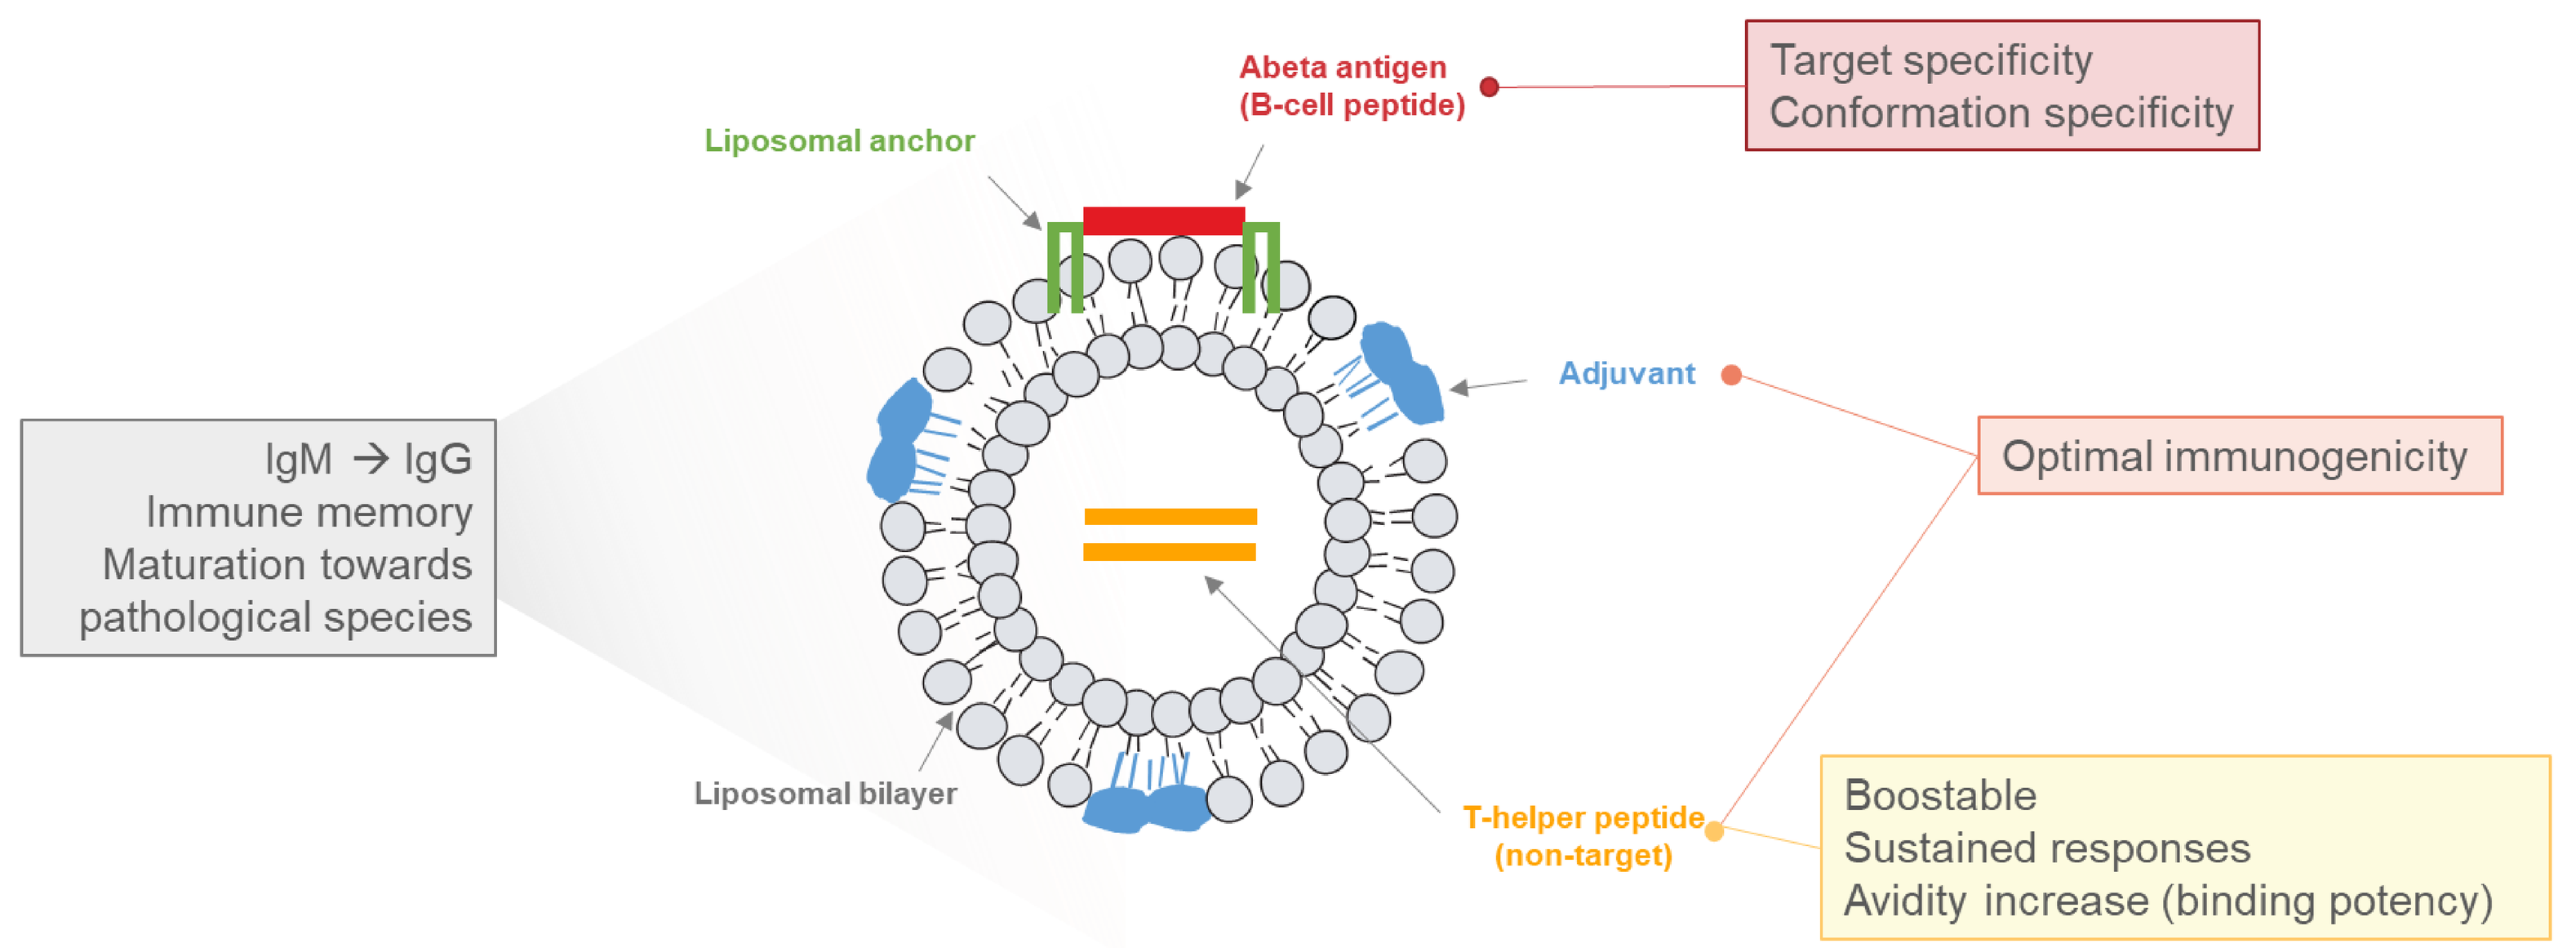
Abeta antigen
(B-cell peptide)
Target specificity
Conformation specificity
Liposomal anchor
Adjuvant
Optimal immunogenicity
IgM → IgG
Immune memory
Maturation towards
pathological species
Liposomal bilayer
T-helper peptide
(non-target)
Boostable
Sustained responses
Avidity increase (binding potency)

1 **Vaccine containing Abeta peptide in a beta-sheet conformation** (designed to raise antibodies against pathological Abeta species)

2 **High and boostable immunogenicity against Abeta** (with NON-Abeta T-helper activation)

3 **Broad epitope coverage of oligomeric and pyro-Glu Abeta** (i.e., the targets of lecanemab and donanemab)

4 **Safe and well tolerated** (no ARIA-E observed as antibody response progressively builds in the body)

Strong recognition of Abeta oligomers and pyroGlu Abeta pathological species

In NHPs, ACI-24.060 induces strong, sustained, boostable IgG response against Abeta oligomers and pyroglutamate Abeta

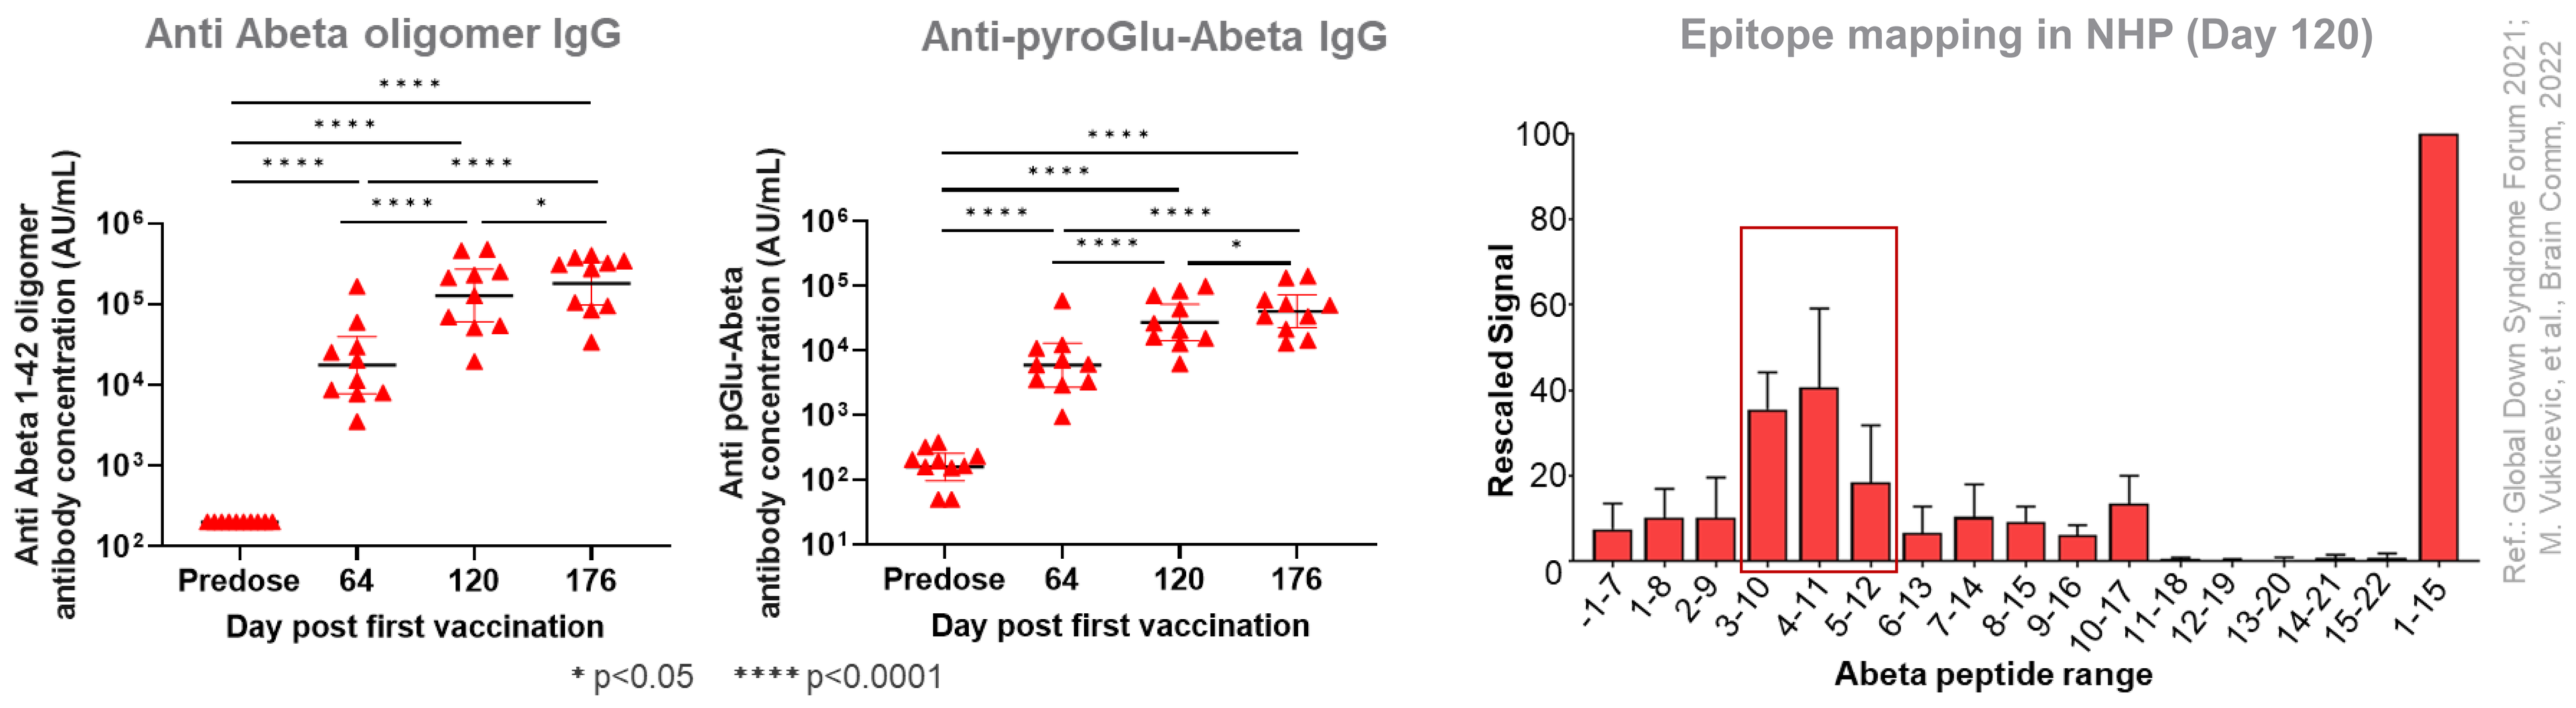
Anti Abeta oligomer IgG
Anti Abeta 1-42 oligomer antibody concentration (AU/mL)
Predose
64
120
176
Day post first vaccination
Anti-pyroGlu-Abeta IgG
Anti pGlu-Abeta antibody concentration (AU/mL)
Epitope mapping in NHP (Day 120)
Rescaled Signal
Abeta peptide range
* p<0.05 **** p<0.0001
Ref.: Global Down Syndrome Forum 2021; M. Vukicevic, et al., Brain Comm, 2022

Fig 2: *Anti-pyroGlu Abeta (left panel) and anti-Abeta oligomer (middle panel) IgG titers in the serum of individual monkeys (n=10) at predose and at Days 64, 120 and 176 after first immunization with ACI-24.060. Data are expressed as individual titers and geometric mean with 95% confidence interval (CI). Anti-Abeta epitope mapping profile of serum samples at Day 120 after the first immunization (right panel). Intensity signals were rescaled from 1 to 100 for each of the four individual monkeys.*

Methods:

Competitive inhibition experiments: assay set-up

In competitive inhibition experiments, sera from NHPs immunized with ACI-24.060 were pre-incubated with increasing concentrations of Abeta monomers or Abeta oligomers in solution before being analyzed in a Meso Scale Discovery (MSD) plate coated with Abeta oligomers (Fig 3). Inhibition was quantified as reduction of the signal as compared to the untreated sample.

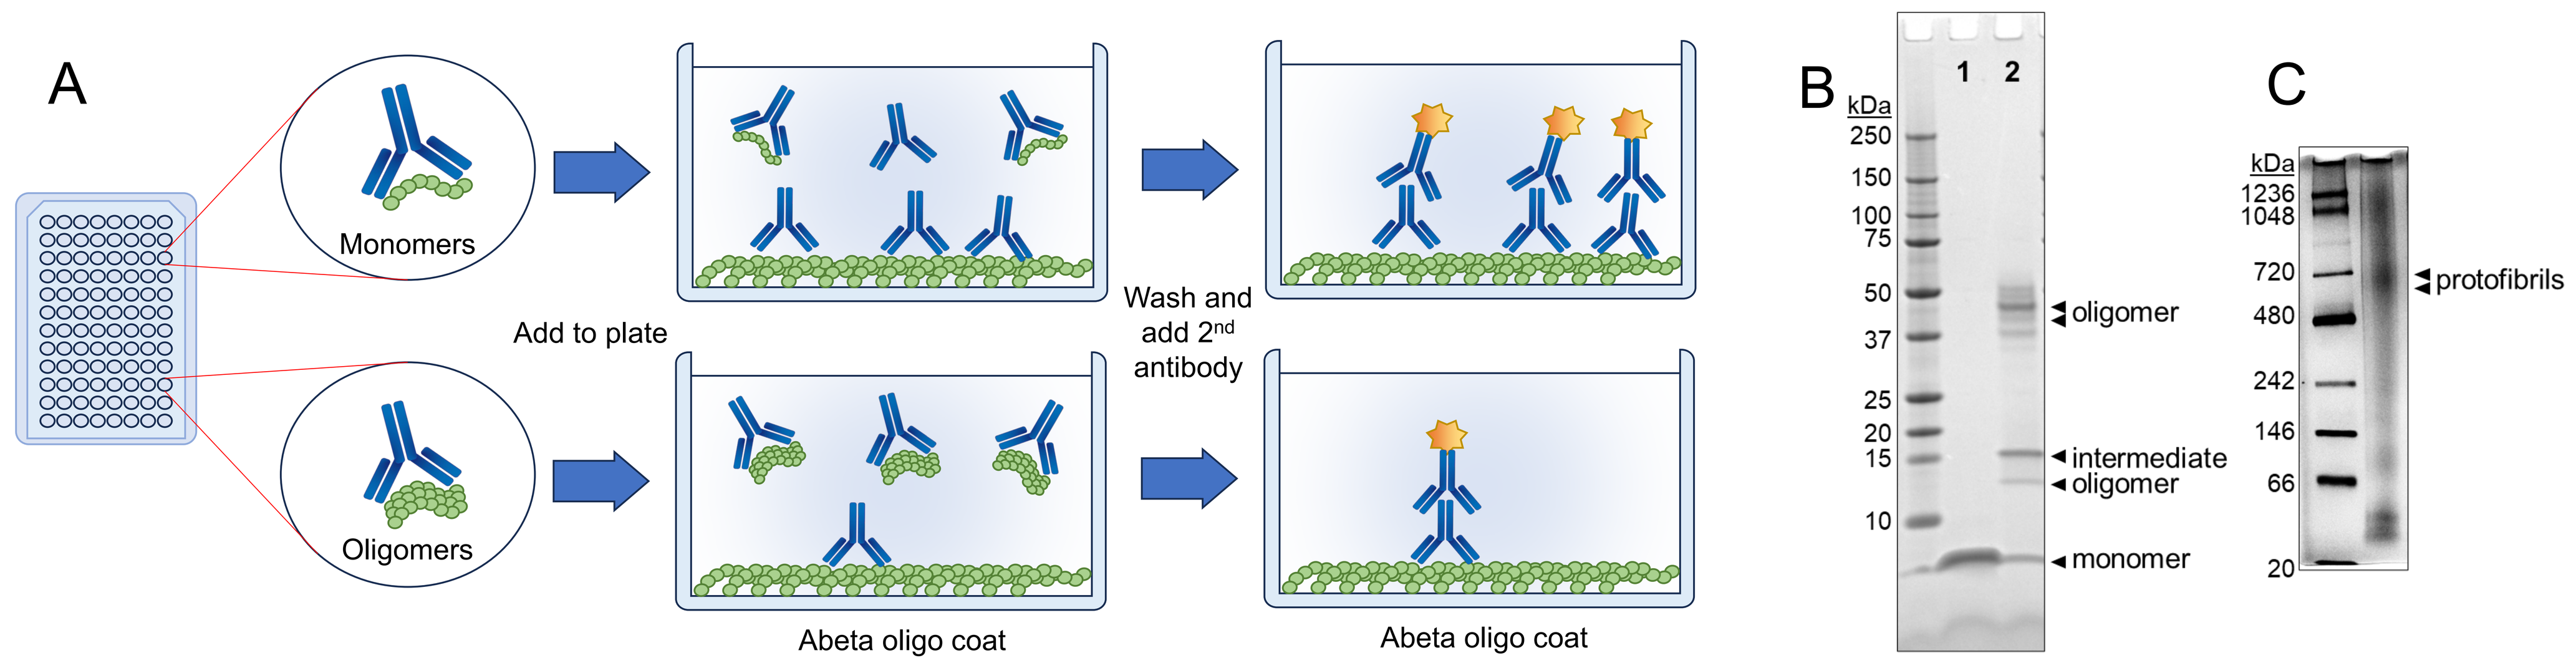
A
Monomers
Oligomers
Add to plate
Wash and add 2nd antibody
Abeta oligo coat
Abeta oligo coat
B
kDa
1 2
oligomer
intermediate oligomer
monomer
C
kDa
protofibrils

***Fig 3**. A:schematic representation of the assay set-up, B: Coomassie stained 10-20% SDS-PAGE of Abeta monomers (well 1) or oligomers (well 2) used in the competitive inhibition experiments, C: Coomassie stained 3-12% PAGE native gel of Abeta oligomers*

In an immunofluorescence (IF) experiment, sera from NHPs immunized with ACI-24.060 were pre-incubated with increasing concentrations of Abeta oligomers in solution before being used to stain Abeta plaques in AD brain sections.

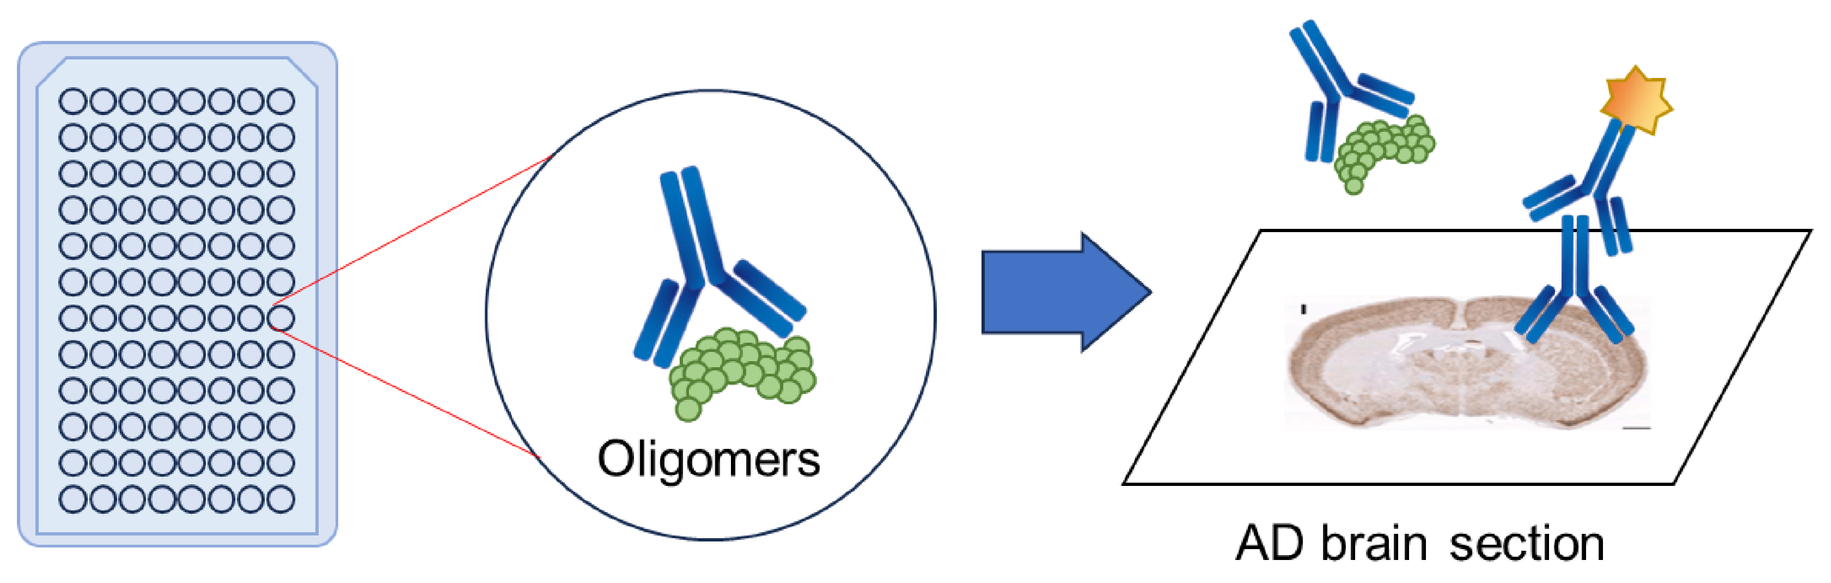
Oligomers
AD brain section

Fig 4: *schematic representation of the assay set-up*

Results:

ACI-24.060 induces antibodies with 3-log preferential recognition of Abeta oligomers vs monomers, similar to lecanemab

In competitive inhibition experiments, IgGs generated by immunization of NHPs with ACI-24.060 gave a 3-log differential for oligomeric over monomeric species of Abeta, similar to lecanemab (Fig 5).

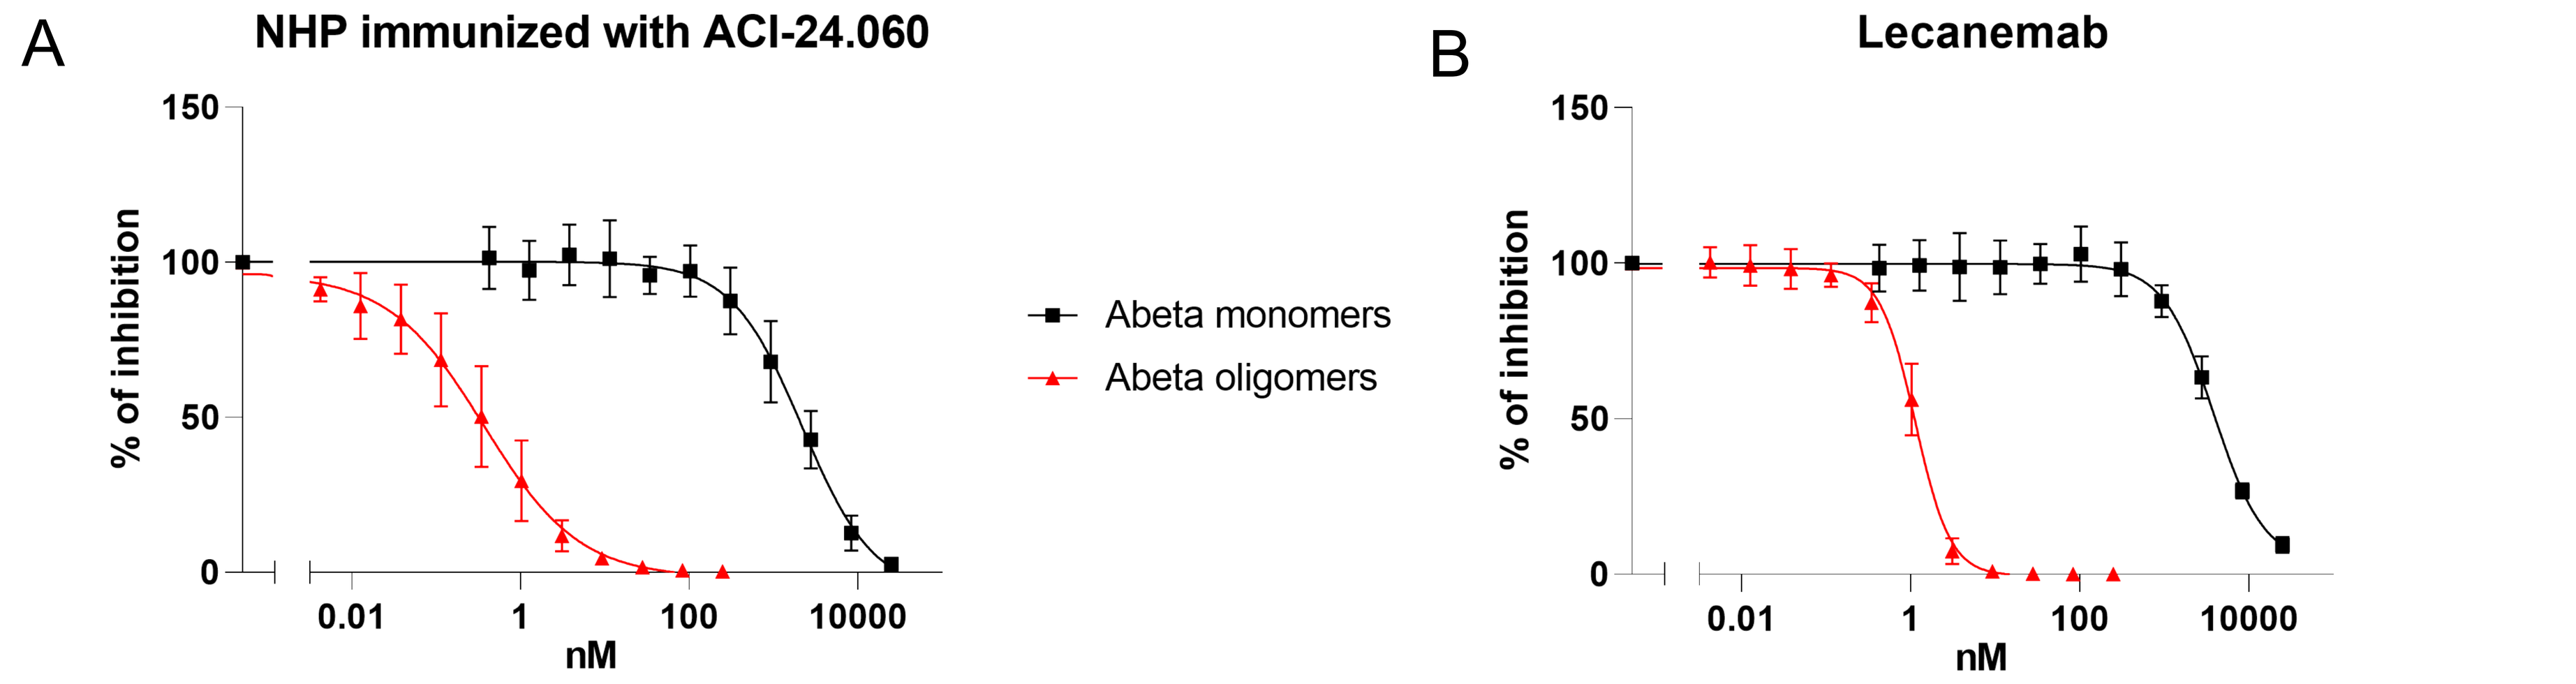
A
NHP immunized with ACI-24.060
% of inhibition
nM
Abeta monomers
Abeta oligomers
B
Lecanemab

NHP at D176 (9 animals)	Min IC50	Max IC50	Average	Ratio Average IC50 Monomers/ Average IC50 Oligomers
IC50 nM Monomers	1217	3192	**2196**	**4733**
IC50 nM Oligomers	0.04	0.98	**0.46**	

Lecanemab (4 repetitions)	Min IC50	Max IC50	Average	Ratio Average IC50 Monomers/ Average IC50 Oligomers
IC50 nM Monomers	3701	4029	**3842**	**3394**
IC50 nM Oligomers	0.74	1.43	**1.13**	

Fig 5: *Nine NHP sera collected after 7 injections with ACI-24.060 at dilution 1/400 (A) and lecanemab at the concentration of 0.5µg/ml (B) were incubated with increasing concentration of Abeta oligomers (triangles) or monomers (squares) and then analyzed in an MSD plate coated with 5µg/mL of Abeta oligomers. The percentage of signal in the presence of Abeta species is normalized against the untreated sample. IC50 of inhibition was calculated and used to obtain the monomer vs oligomer ratio. Graph in A shows the inhibition mean ±SD of nine NHPs, while B shows the inhibition mean ±SD of four independent experiments.*

Pre-incubation of immune NHP sera with Abeta oligomers blocks AD plaque staining

IgGs generated by immunization of NHPs with ACI-24.060 recognize Abeta plaques in brains of AD donors. Pre-incubation with Abeta oligomers, but not with scrambled Abeta peptide (not shown), abolishes plaque recognition in a dose-dependent manner (Fig 6).

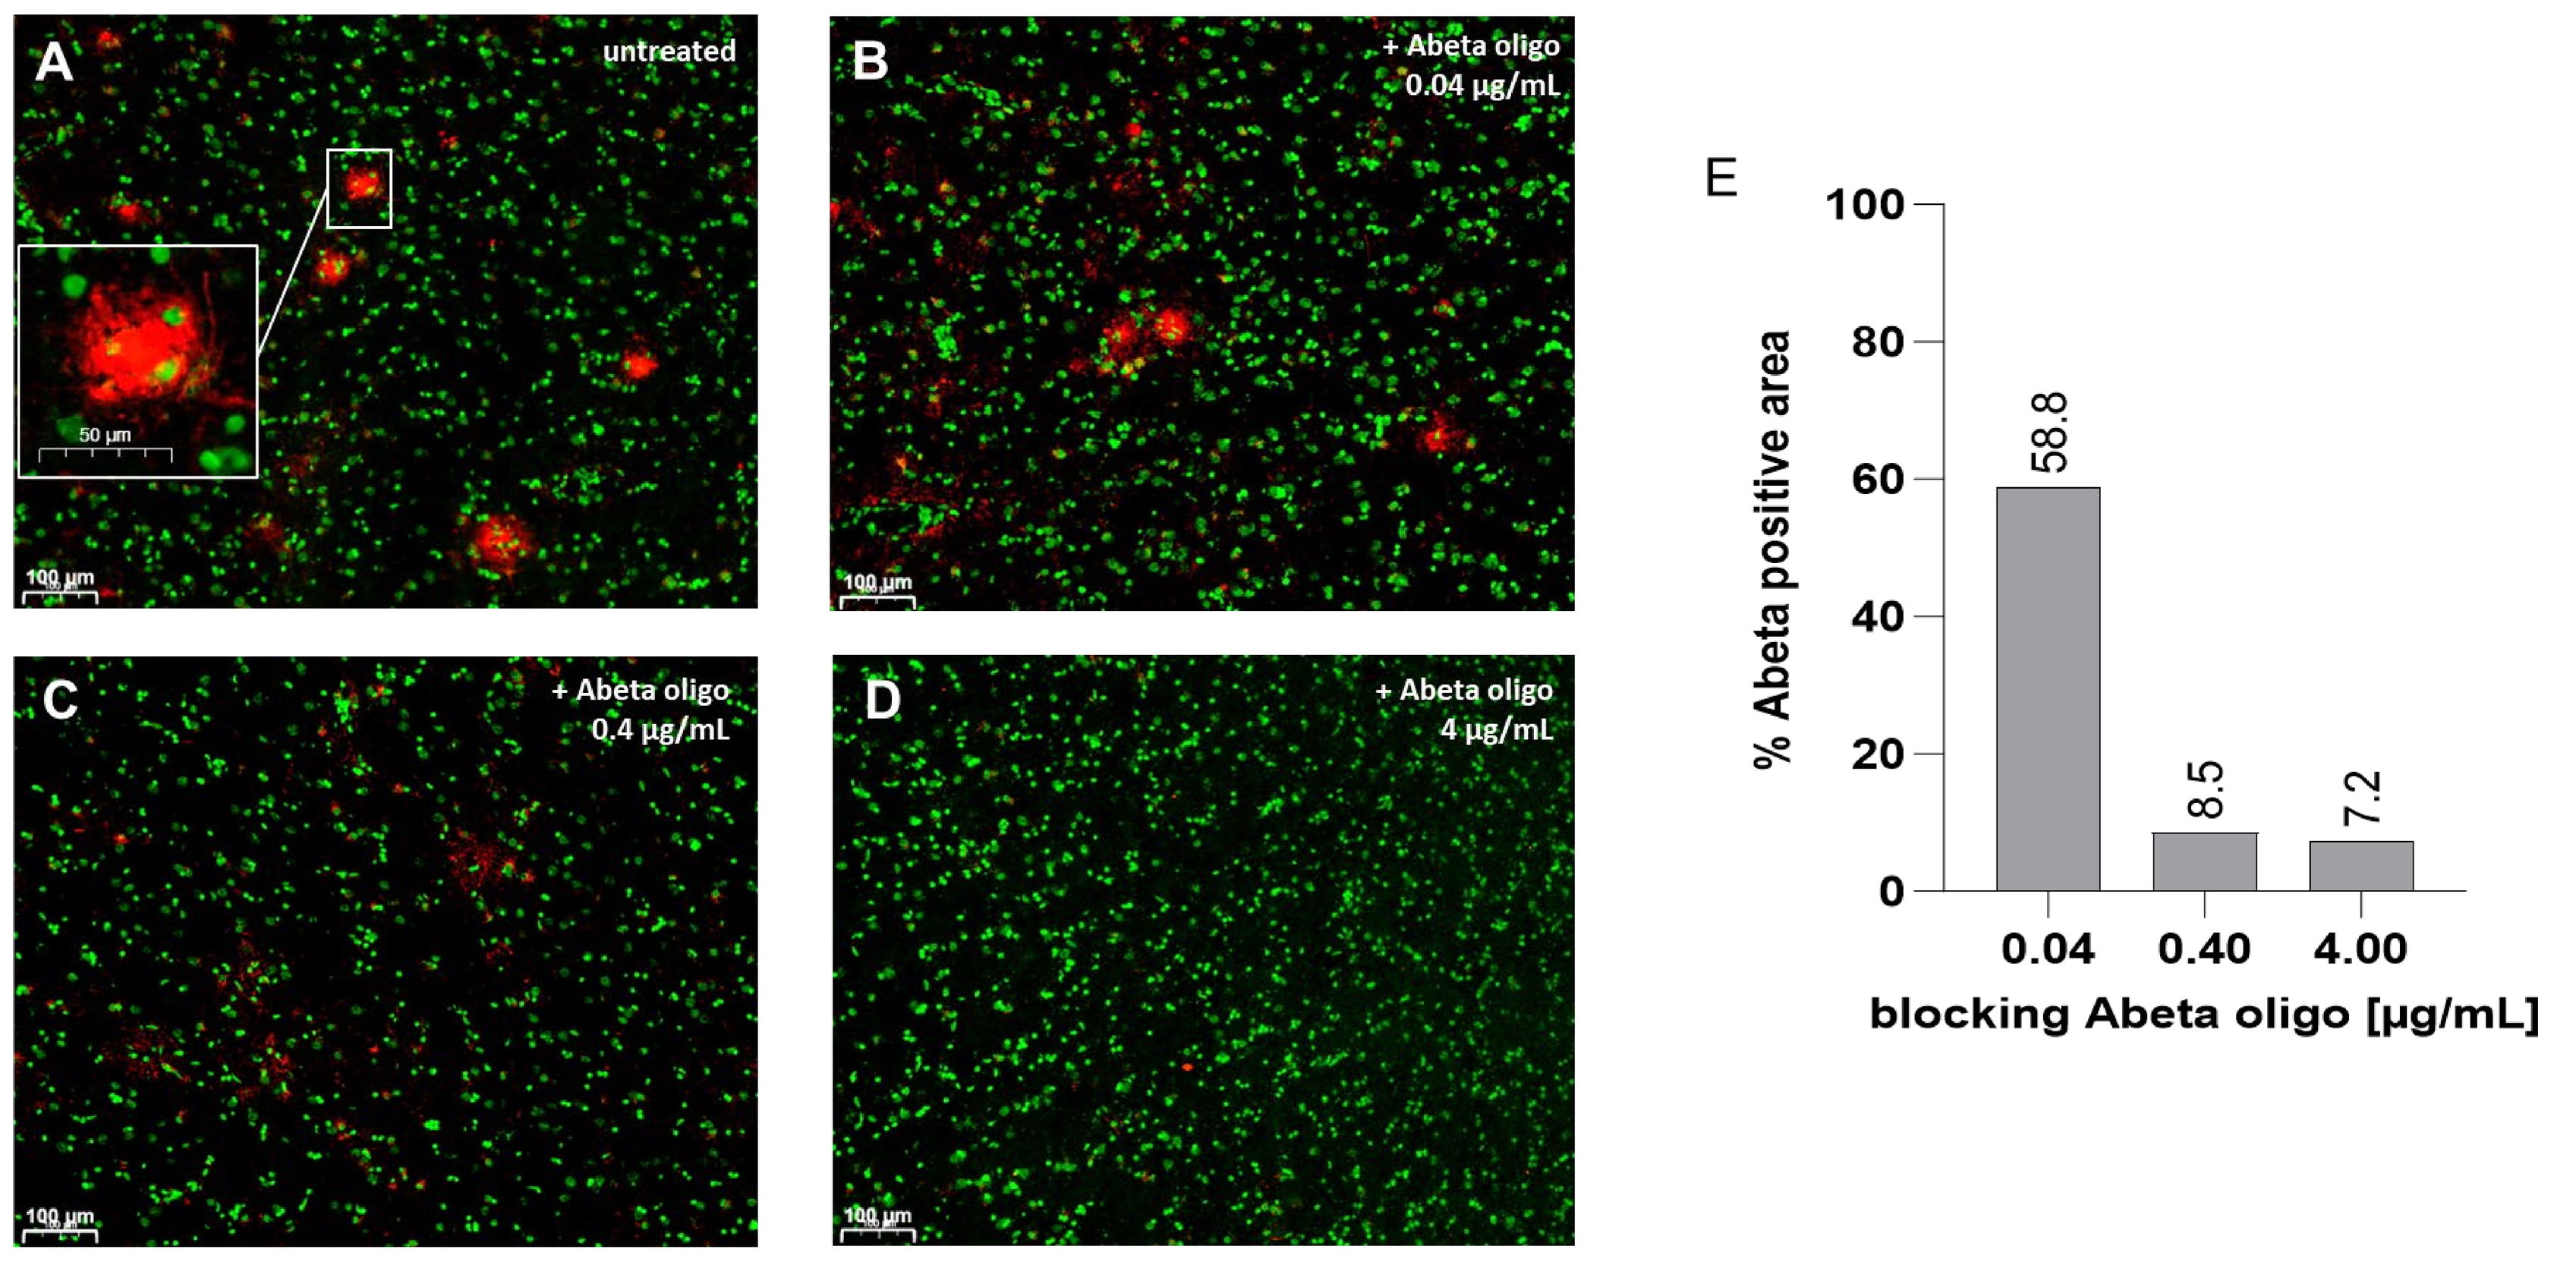
A
untreated
B
+ Abeta oligo 0.04 µg/mL
C
+ Abeta oligo 0.4 µg/mL
D
+ Abeta oligo 4 µg/mL
E
% Abeta positive area
58.8
8.5
7.2
0.04
0.40
4.00
blocking Abeta oligo [µg/mL]

Fig 6: *NHP sera collected after 7 injections with ACI-24.060 were pre-incubated with increasing concentrations of Abeta oligomers (B-D) before being used to stain Abeta plaques in frozen AD brain sections (DAPI in green, Abeta in red). The graph (E) shows the percentage of Abeta positive area recognized by sera pre-treated with Abeta oligomers normalized to the untreated serum*

PyroGlu Abeta recognition of immune NHP sera similar to donanemab

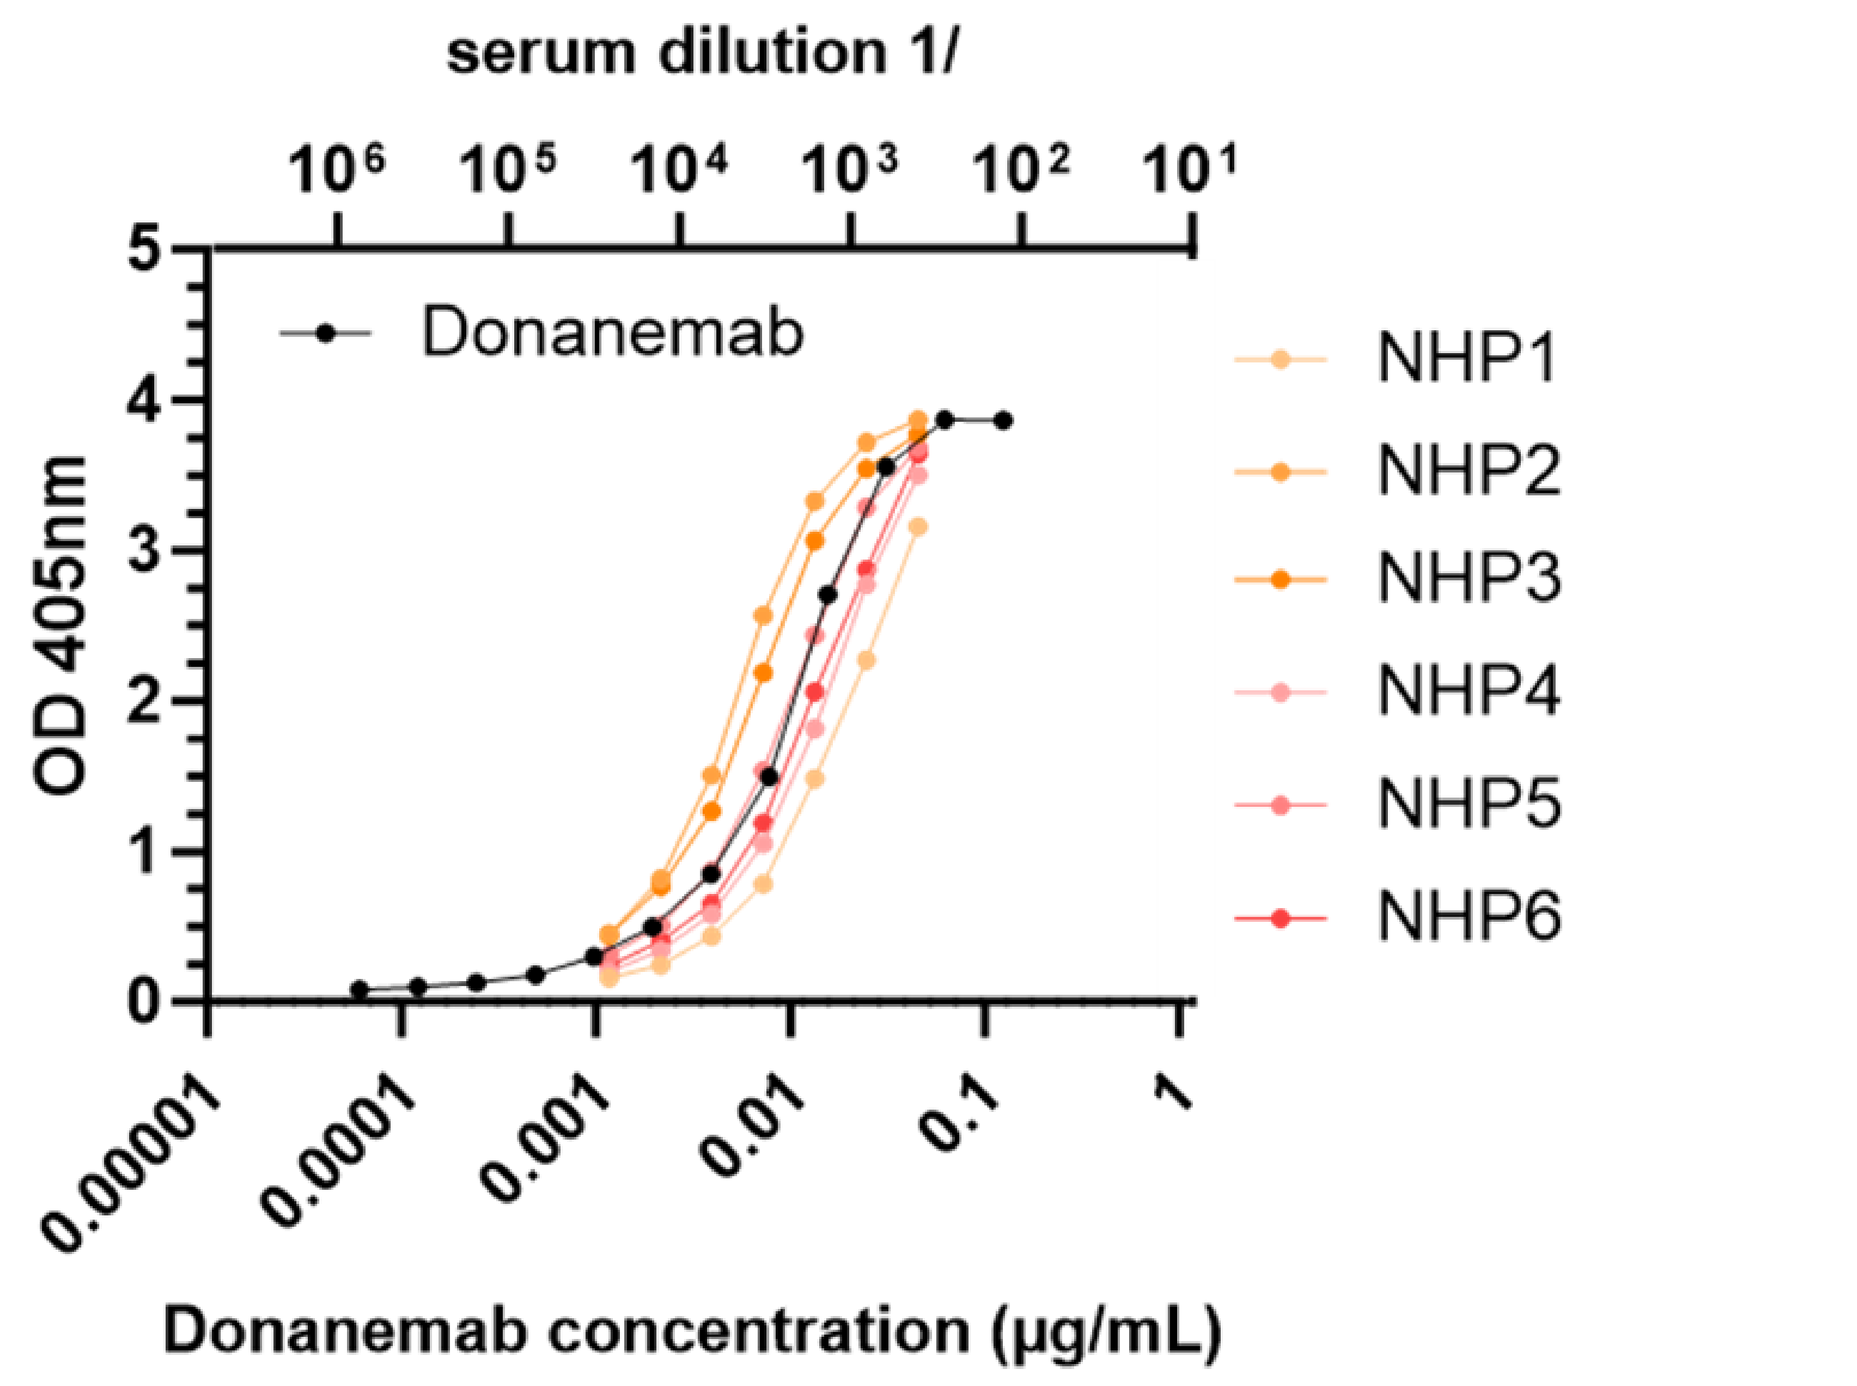
serum dilution 1/
OD 405nm
Donanemab
NHP1
NHP2
NHP3
NHP4
NHP5
NHP6
Donanemab concentration (µg/mL)

***Fig 7**: Anti-pyroGlu-Abeta IgG ELISA. Six NHP sera collected after 7 injections with ACI-24.060 were tested in parallel with donanemab. Eight 2-fold dilutions from an initial 100-fold dilution or 0.25µg/mL for the sera (upper x-axis) or donanemab (lower x-axis) respectively were plotted against OD values shown on y-axis.*

Conclusions:

ACI-24.060 induced antibody responses in NHPs with:

- Strong binding to pathological Abeta oligomers and pyroGlu-Abeta species
- 3-log preference in binding to oligomeric versus monomeric Abeta, similar to lecanemab
- AD brain plaque recognition that could be blocked by pre-incubation with Abeta oligomers
- Recognition of pyroGlu-Abeta in the range of binding of clinically relevant concentrations of donanemab

Altogether, these data confirmed that ACI-24.060 induces robust levels of anti- Abeta antibodies, with an excellent binding profile, combining the properties of lecanemab and donanemab.

CTAD 2023, Boston